88th EAS Congress, 2020

Treatment patterns and healthcare resource use in primary hypercholesterolaemia and mixed dyslipidaemia: results of a UK Delphi panel

Llewellyn S[1], Bilitou A[2], Dunton K[3], Åkesson C[1], Hall K[1], Quinn C[1]

[1]Vitaccess Ltd, Oxford, UK; [2]Daiichi Sankyo Europe GmbH, Munich, Germany; [3]Daiichi Sankyo UK Ltd, Gerrards Cross, London, UK

Background

- Mendelian randomisation, prospective epidemiologic cohort studies, randomised clinical trials and meta-analyses have shown that elevated low-density lipoprotein cholesterol (LDL-C) is a direct causal factor of atherosclerotic cardiovascular disease (ASCVD).[1-2]
- ASCVD is a leading cause of death and morbidity in Europe and the UK.[3-4]
- The importance of lowering LDL-C to prevent CVD is strongly emphasised in treatment guidelines in Europe by the ESC/EAS[5] (see Figure 1) as well as in the UK by NICE Clinical Guidance.[6]

Aim

To elicit estimates on the contemporary treatment patterns for patients treated for hypercholesterolaemia/mixed dyslipidaemia in the UK.

Figure 1: Treatment algorithm for LDL-C lowering according to guidelines[3-4]

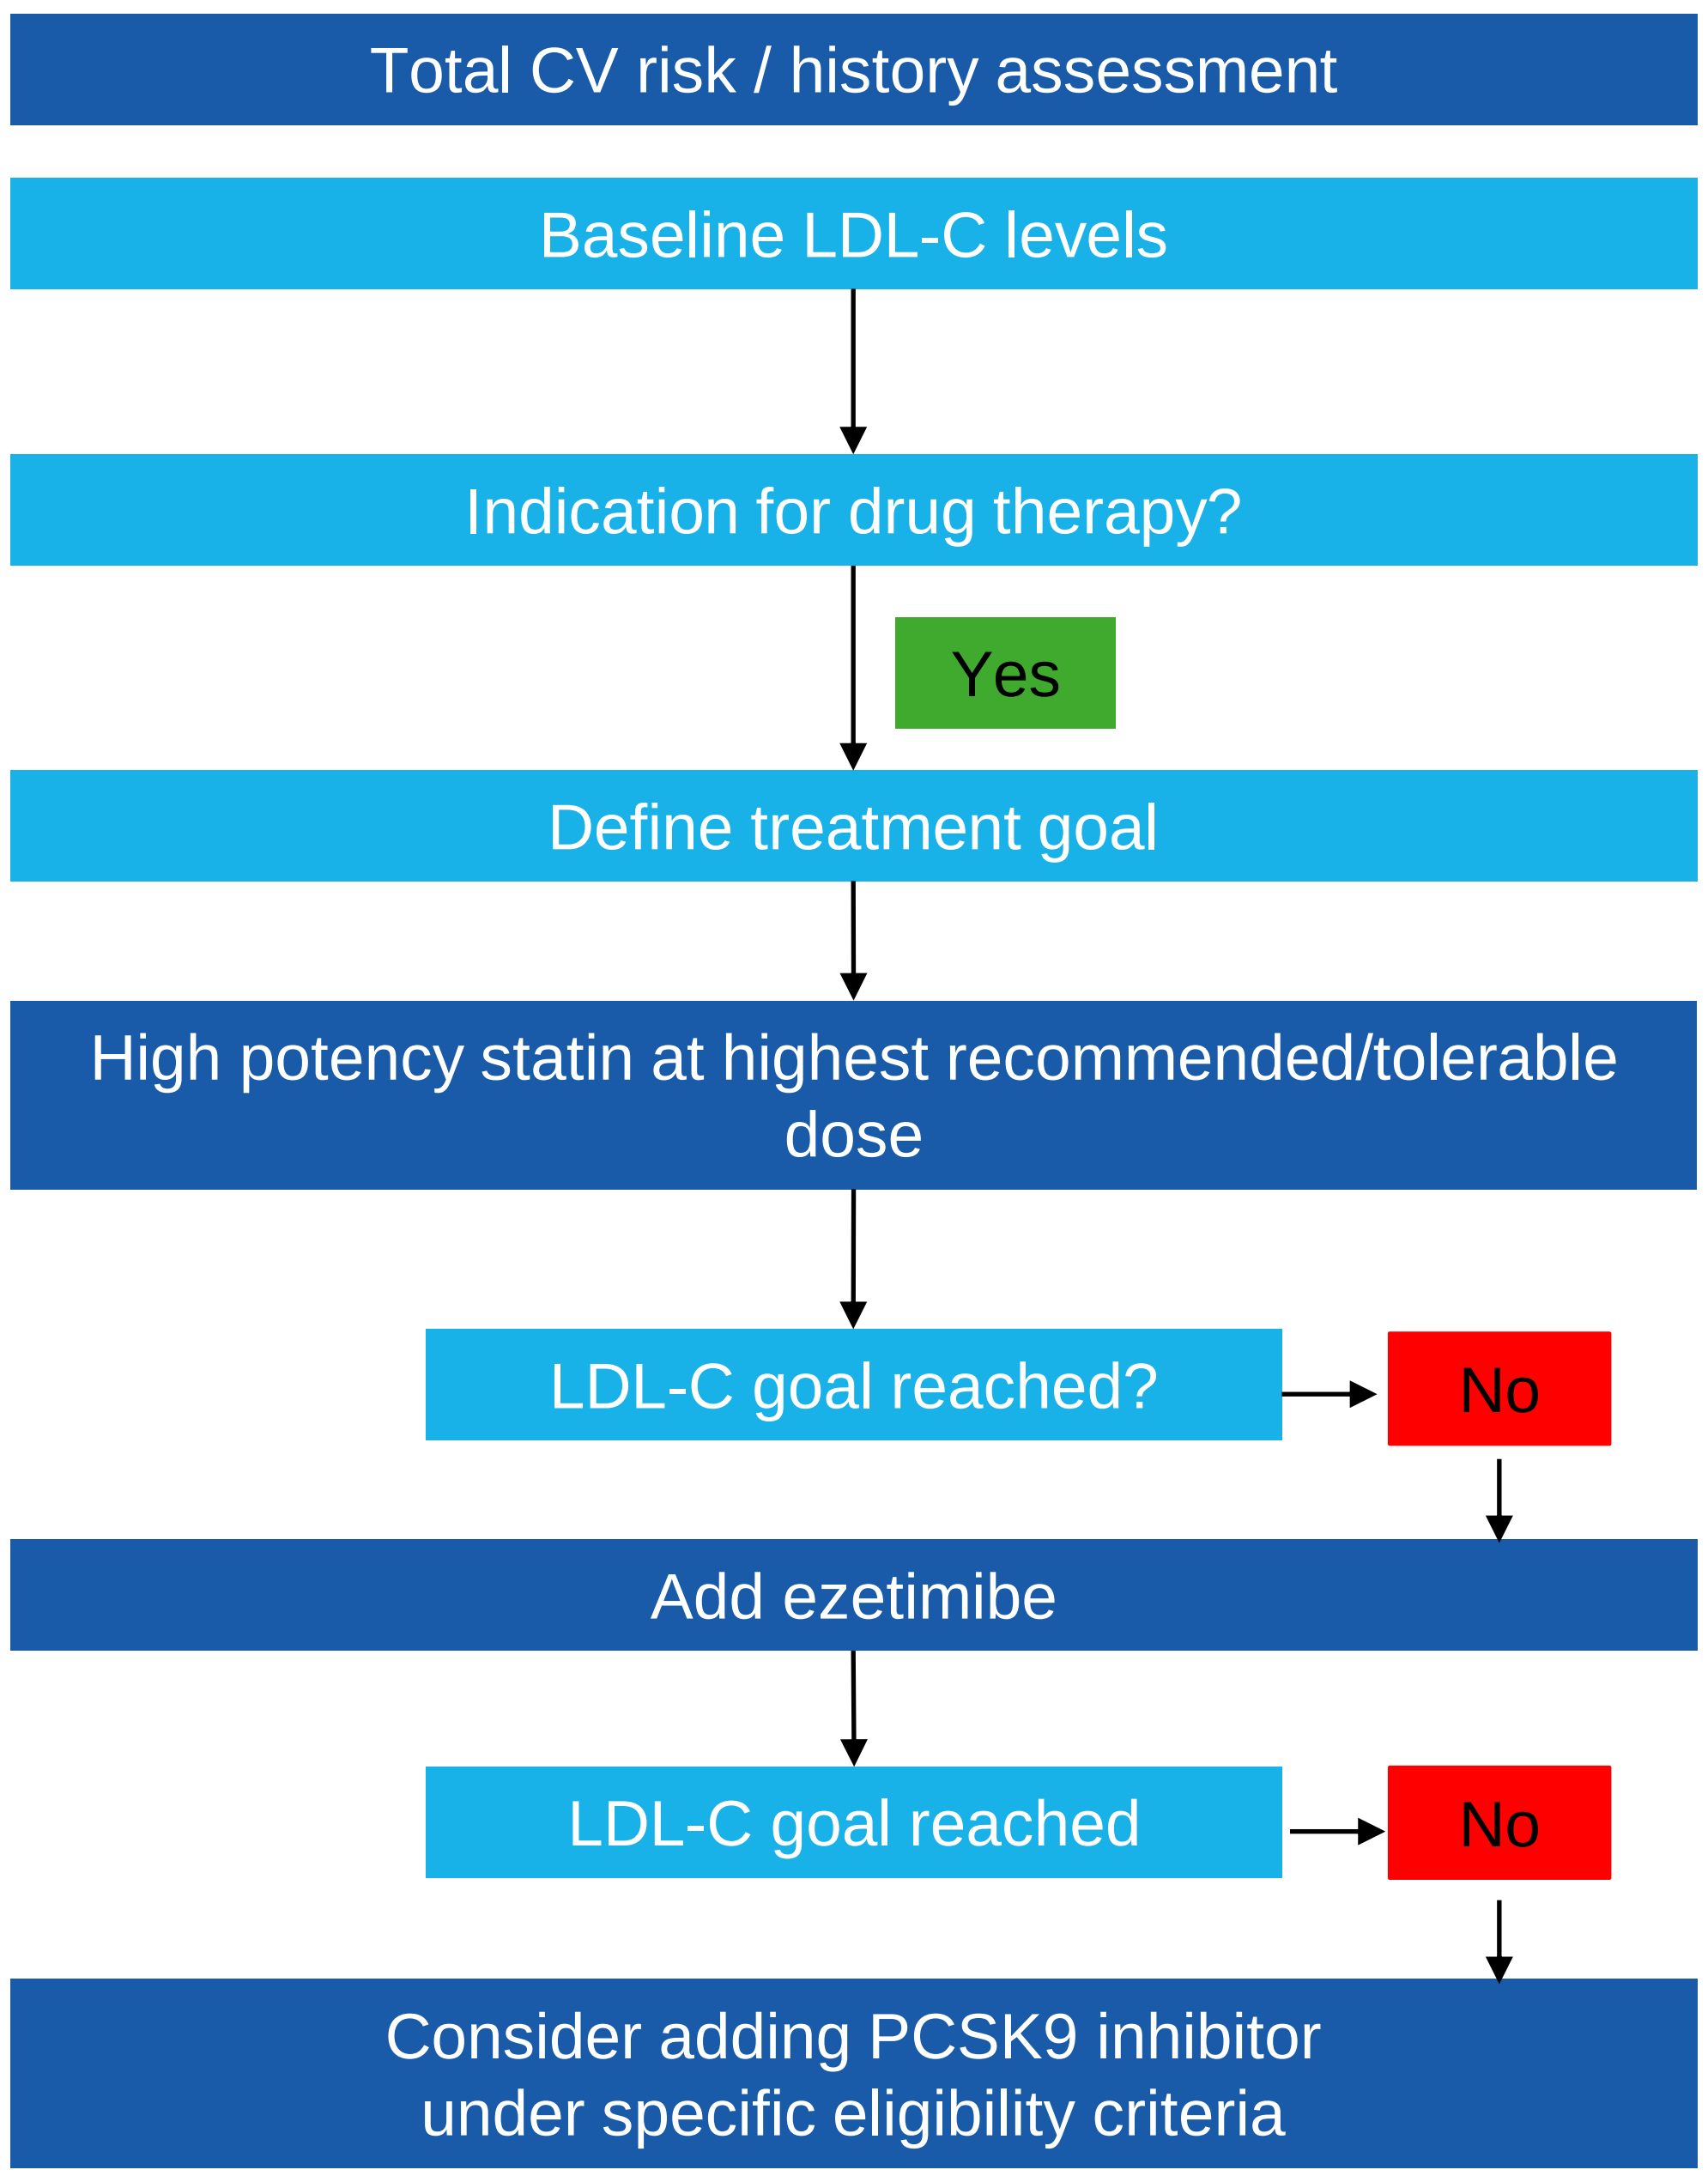
Total CV risk / history assessment
Baseline LDL-C levels
Indication for drug therapy?
Yes
Define treatment goal
High potency statin at highest recommended/tolerable dose
LDL-C goal reached?
No
Add ezetimibe
LDL-C goal reached
No
Consider adding PCSK9 inhibitor under specific eligibility criteria

Adapted from Mach et al., 2020[5]

Methods

- The Delphi methodology was used to elicit responses in lipid management; this is a widely accepted method for achieving convergence of opinion concerning real-world knowledge solicited from experts.[7] A feedback iterative process is used to allow and encourage panel participants to reassess, refine and modify their initial judgements to reach consensus.
- Experts from the UK were recruited using a screener with pre-specified inclusion criteria:
 - >5 years experience in treating patients with hypercholesterolaemia
 - >30 patients seen each year and
 - using LLTs including statins, ezetimibe and/or PCSK9is.
- Via email participants completed a structured questionnaire on the treatment algorithm (Round 1). A web-based moderated Delphi panel was then conducted during which aggregate responses were discussed and voted on in instances where consensus had not been reached (Round 2).
- Anonymised responses were analysed independently using descriptive statistics to provide aggregate estimates (mean and median, % of proportions of patients etc).

Results

Eight experts of different specialties and location in the UK (see Figure 2) completed the questionnaire and seven of them iterated in Round 2 to produce the final results. Aggregated mean estimates provided by experts on how patients with primary hypercholesterolaemia and mixed dyslipidaemia are treated with existing LLTs based on their experience are shown.

Figure 2: Sample of Delphi panel and results as aggregate and anonymised responses (mean estimates are shown)

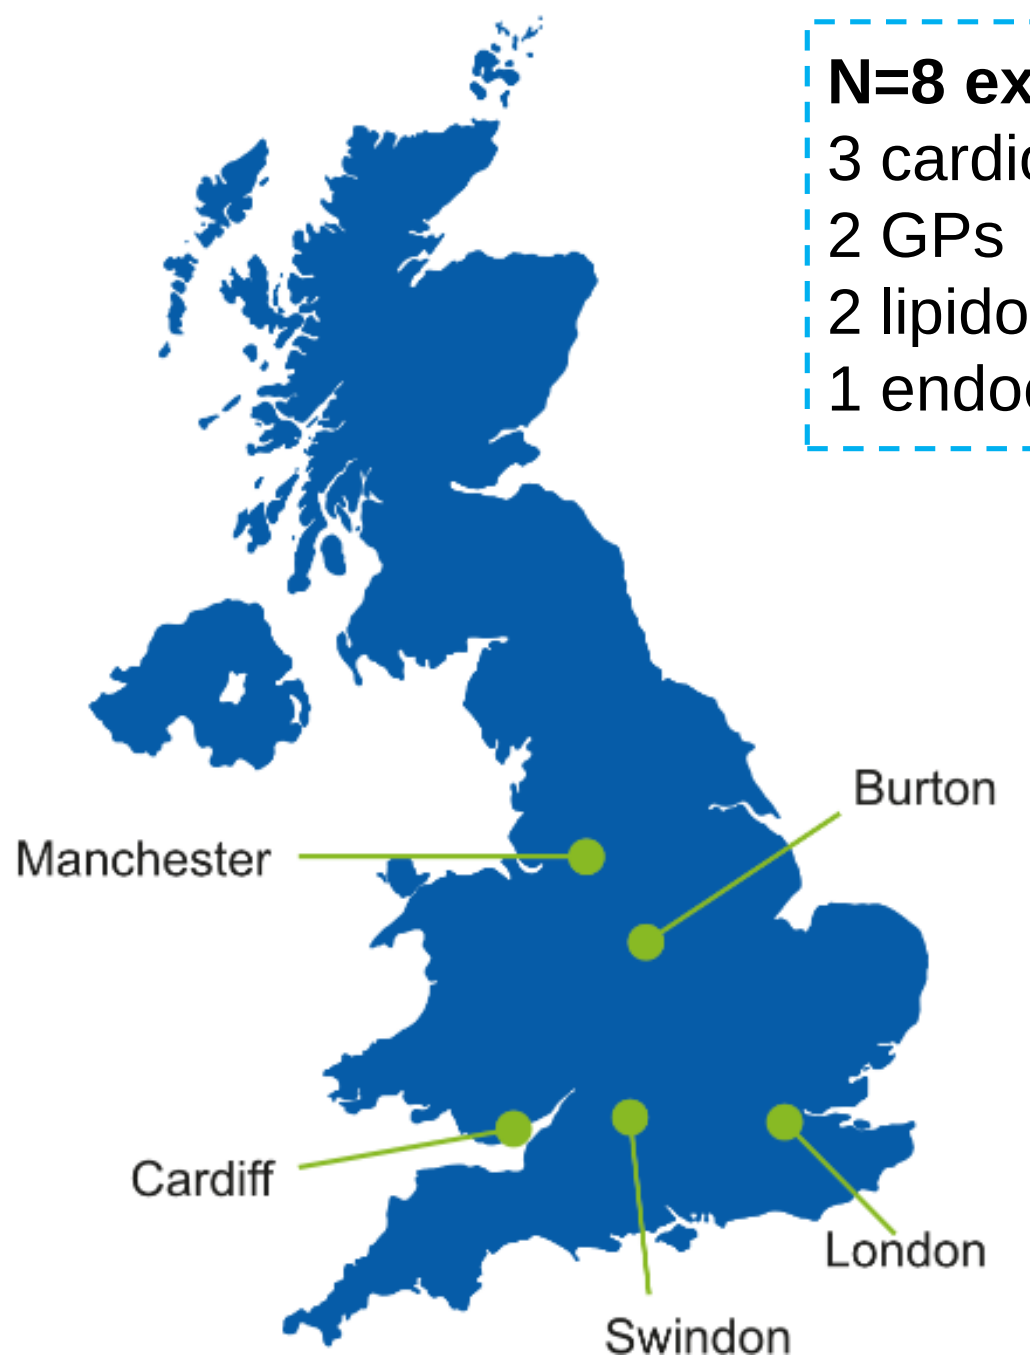
Manchester
Burton
Cardiff
Swindon
London

N=8 experts
3 cardiologists
2 GPs
2 lipidologists
1 endocrinologist

- 17 mean years of experience, range 10-27
- About 1,035 patients seen per year (mean), range >50 -3,000

Estimated mean % of patients seen as reported by experts

Patients with primary hypercholesterolaemia/mixed dyslipidaemia
- 49% presence of CVD
- 15% HeFH
- 13% statin intolerance
- 35% severe hypercholesterolaemia

Among patients with hypercholesterolaemia and mixed dyslipidaemia treated with LLTs
- 50% considered high or very high CV risk

- 81% receive statins (any dose, intensity alone or in combination)
- 7% receive a fixed-dose combination

15% considered **statin intolerant** or contra-indicated to statins
- 35% receive ezetimibe, of whom 64% are considered not at goal
- 9% receive PCSK9 inhibitor

29% of those on **maximally tolerated statin** have not adequate LDL-C control
- 51% receive maximally tolerated statin + ezetimibe of whom 24% are not at goal

Experts noted:

- a mean of 5% of patients were estimated to discontinue statins or ezetimibe per year and 7% of patients on PCSK9 inhibitors.
- it is reasonable to assume that the monitoring (blood tests, consultations) is similar across oral treatments such as statins and ezetimibe; one third mentioned that PCSK9 inhibitors consume more time for monitoring and prescriptions dependent on clinic and set by guidelines.

Overall, the panel confirmed heterogeneity in clinical practice depending on patient characteristics and treatment setting, thus difficulty in defining a “UK average”.

Conclusions

Consensus between experts was difficult to reach due to heterogeneity. Considerations should be taken around all patient and disease-related factors.

Use of PCSK9 inhibitors was reportedly low among patients not at goal and associated with additional administrative burden compared to oral lipid-lowering treatments.

Unmet need still exists in patients at high and very high risk of CVD to appropriately bring LDL-C levels closer to therapeutic goals. For these patients, combination therapy is a reasonable step and consistent with clinical guideline recommendations.

References: [1]Borén et al. (2020) Eur Heart J 41(24): 2313–2330. [2]Ference et al. (2017) Eur Heart J 38(32): 2459–2472. [3]EHN (2017) European Cardiovascular Disease Statistics 2017. Available: http://www.ehnheart.org/cvd-statistics.html. [4]British Heart Foundation (2020) UK Factsheet. Available: https://www.bhf.org.uk/-/media/files/research/heart-statistics/bhf-cvd-statistics-uk-factsheet.pdf?la=en. [5]Mach et al. (2020) 2019 ESC/EAS Guidelines for the management of dyslipidaemias Eur Heart J; 41(1): 111–188. [6]NICE (2016) CG181: Cardiovascular disease: risk assessment and reduction, including lipid modification. Available at: http://www.nice.org.uk/guidance/cg181. [7]Hsu & Sandford (2007). PARE; 12(1): 10.

This study was funded by Daiichi Sankyo Europe.